Physicians' perceptions and attitudes towards digitalization and new technologies in diabetes care

Bernhard Kulzer[1], Timm Roos[1], Dominic Ehrmann[1], Norbert Hermanns[1], Lutz Heinemann[2]

[1]FIDAM - Research Institute Diabetes Academy, Diabetes Center Mergentheim, Bad Mergentheim, Germany,
[2]Science Consulting in Diabetes, Neuss, Germany

ABSTRACT

Digitalization is an emerging field. However, little is known about the perceptions and expectations of physicians towards digitalization and new technologies in diabetes care.

We conducted a survey in secondary care practices throughout Germany. A total of 422 physicians completed the online survey (46% female; mean age: 53 years; mean work experience: 25 years). Per practice, 160 people with diabetes used Flash sensor-based glucose monitoring, 41 used real-time continuous glucose monitoring, and 60 used insulin pump therapy. 79.6% of physicians reported a positive or very positive attitude towards digitalization in diabetes care which was somewhat higher than for general digitalization (63.7% reported a positive or very positive attitude).

Major advantages of digitalization referred to patient safety (63.4%), better quality of care (62.4%), and decision support (61.9%). The three major barriers were costs (54.6%), legal issues (49.2%), and concerns about data security (40.3%). Current and expected use in 5 years of new technologies are shown in the figure.

The potential for digitalization in diabetes is perceived as very high by physicians. However, costs and safety issues need to be addressed to overcome barriers towards implementation of digital solutions. Currently, the use of digital solutions in routine practice is not common and is expected to grow only marginally in the next 5 years.

INTRODUCTION

Diabetes is a "data-driven condition" and new diabetes technologies are introduced into routine care. Therefore, there are many opportunities how digital applications and technologies can influence diabetes care – for physicians/healthcare providers and people with diabetes. However, there are also potential risks and barriers towards an increasing digitalization of healthcare.

In this analysis, we wanted to shed some light on the perceptions and expectations of physicians towards digitalization and new technologies in diabetes care.

METHODS

We conducted a survey in secondary care practices throughout Germany. A total of 422 physicians completed a questionnaire regarding different aspects of digitalization and diabetes technology online (46% female; mean age: 53 years; mean work experience: 25 years; 54.3% in rural areas).

All answers reflect the appraisals or estimates of participating physicians.

RESULTS

Use of diabetes technology

- In the participating practices, a total of 67,769 people with type 1 and type 2 diabetes are currently using Flash Glucose Monitoring, followed by 25,007 insulin pump users, and 17,169 real-time continuous glucose monitoring (CGM) users (Figure 1).
 - o Thus, per practice, 160 people with diabetes used Flash Glucose Monitoring, 41 used real-time CGM, and 60 used insulin pump therapy.
- Interestingly, the absolute number of Flash Glucose Monitoring users is nearly identical for people with type 1 and type 2 diabetes.

Attitudes towards digitalization

- 79.6% of physicians reported a positive or very positive attitude towards digitalization in diabetes care (Figure 2). Only 17.3% reported a negative attitude.
- This was somewhat higher than their attitude towards digitalization in general (63.7% reported a positive or very positive attitude; Figure 3).

Advantages and barriers of digitalization

- Major advantages of digitalization (Figure 4) referred to patient safety (63.4%), better quality of care (62.4%), and decision support (61.9%). Greater job satisfaction (33.8%) and better economic efficiency were not as strongly regarded as an advantage (28.4%).
- The three major barriers towards digitalization (Figure 5) were the high investment costs (54.6%), legal uncertainties (49.2%), and concerns about data security (40.3%). Risk of mistreatment was not strongly considered a barrier (11.9%).

Current and future use of digital diabetes care (Figure 6)

- Physicians estimated that only 28.2% of their patients are currently using a glucose management software and that in 5 years this will rise to a total of 49.4%.
- Interestingly, a similar estimate was given regarding the use of diabetes apps.
- Based on physician's estimate, digital education and telemedical consulting will only reach 35% and 21% in 5 years, respectively.

DISCUSSION

Overall, physicians have a very positive attitude towards digitalization in diabetes care that also exceeds their attitude towards digitalization in general. Patient-related aspects of digitalization were considered as more advantageous than personal-related aspects.

However, personal-related aspects such as high investment costs as well as legal and safety issues were considered as important barriers and need to be addressed to overcome barriers towards implementation of digital solutions.

Currently, the use of digital solutions in routine practice is not common and is expected to grow only marginally in the next 5 years. Especially the use of telemedical consultations and digital forms of patient education were not rated as reaching many patients. Whether this is due to actual reservations of physicians or expected reservations of patients needs to be further investigated.

Supported by Berlin-Chemie AG, Germany

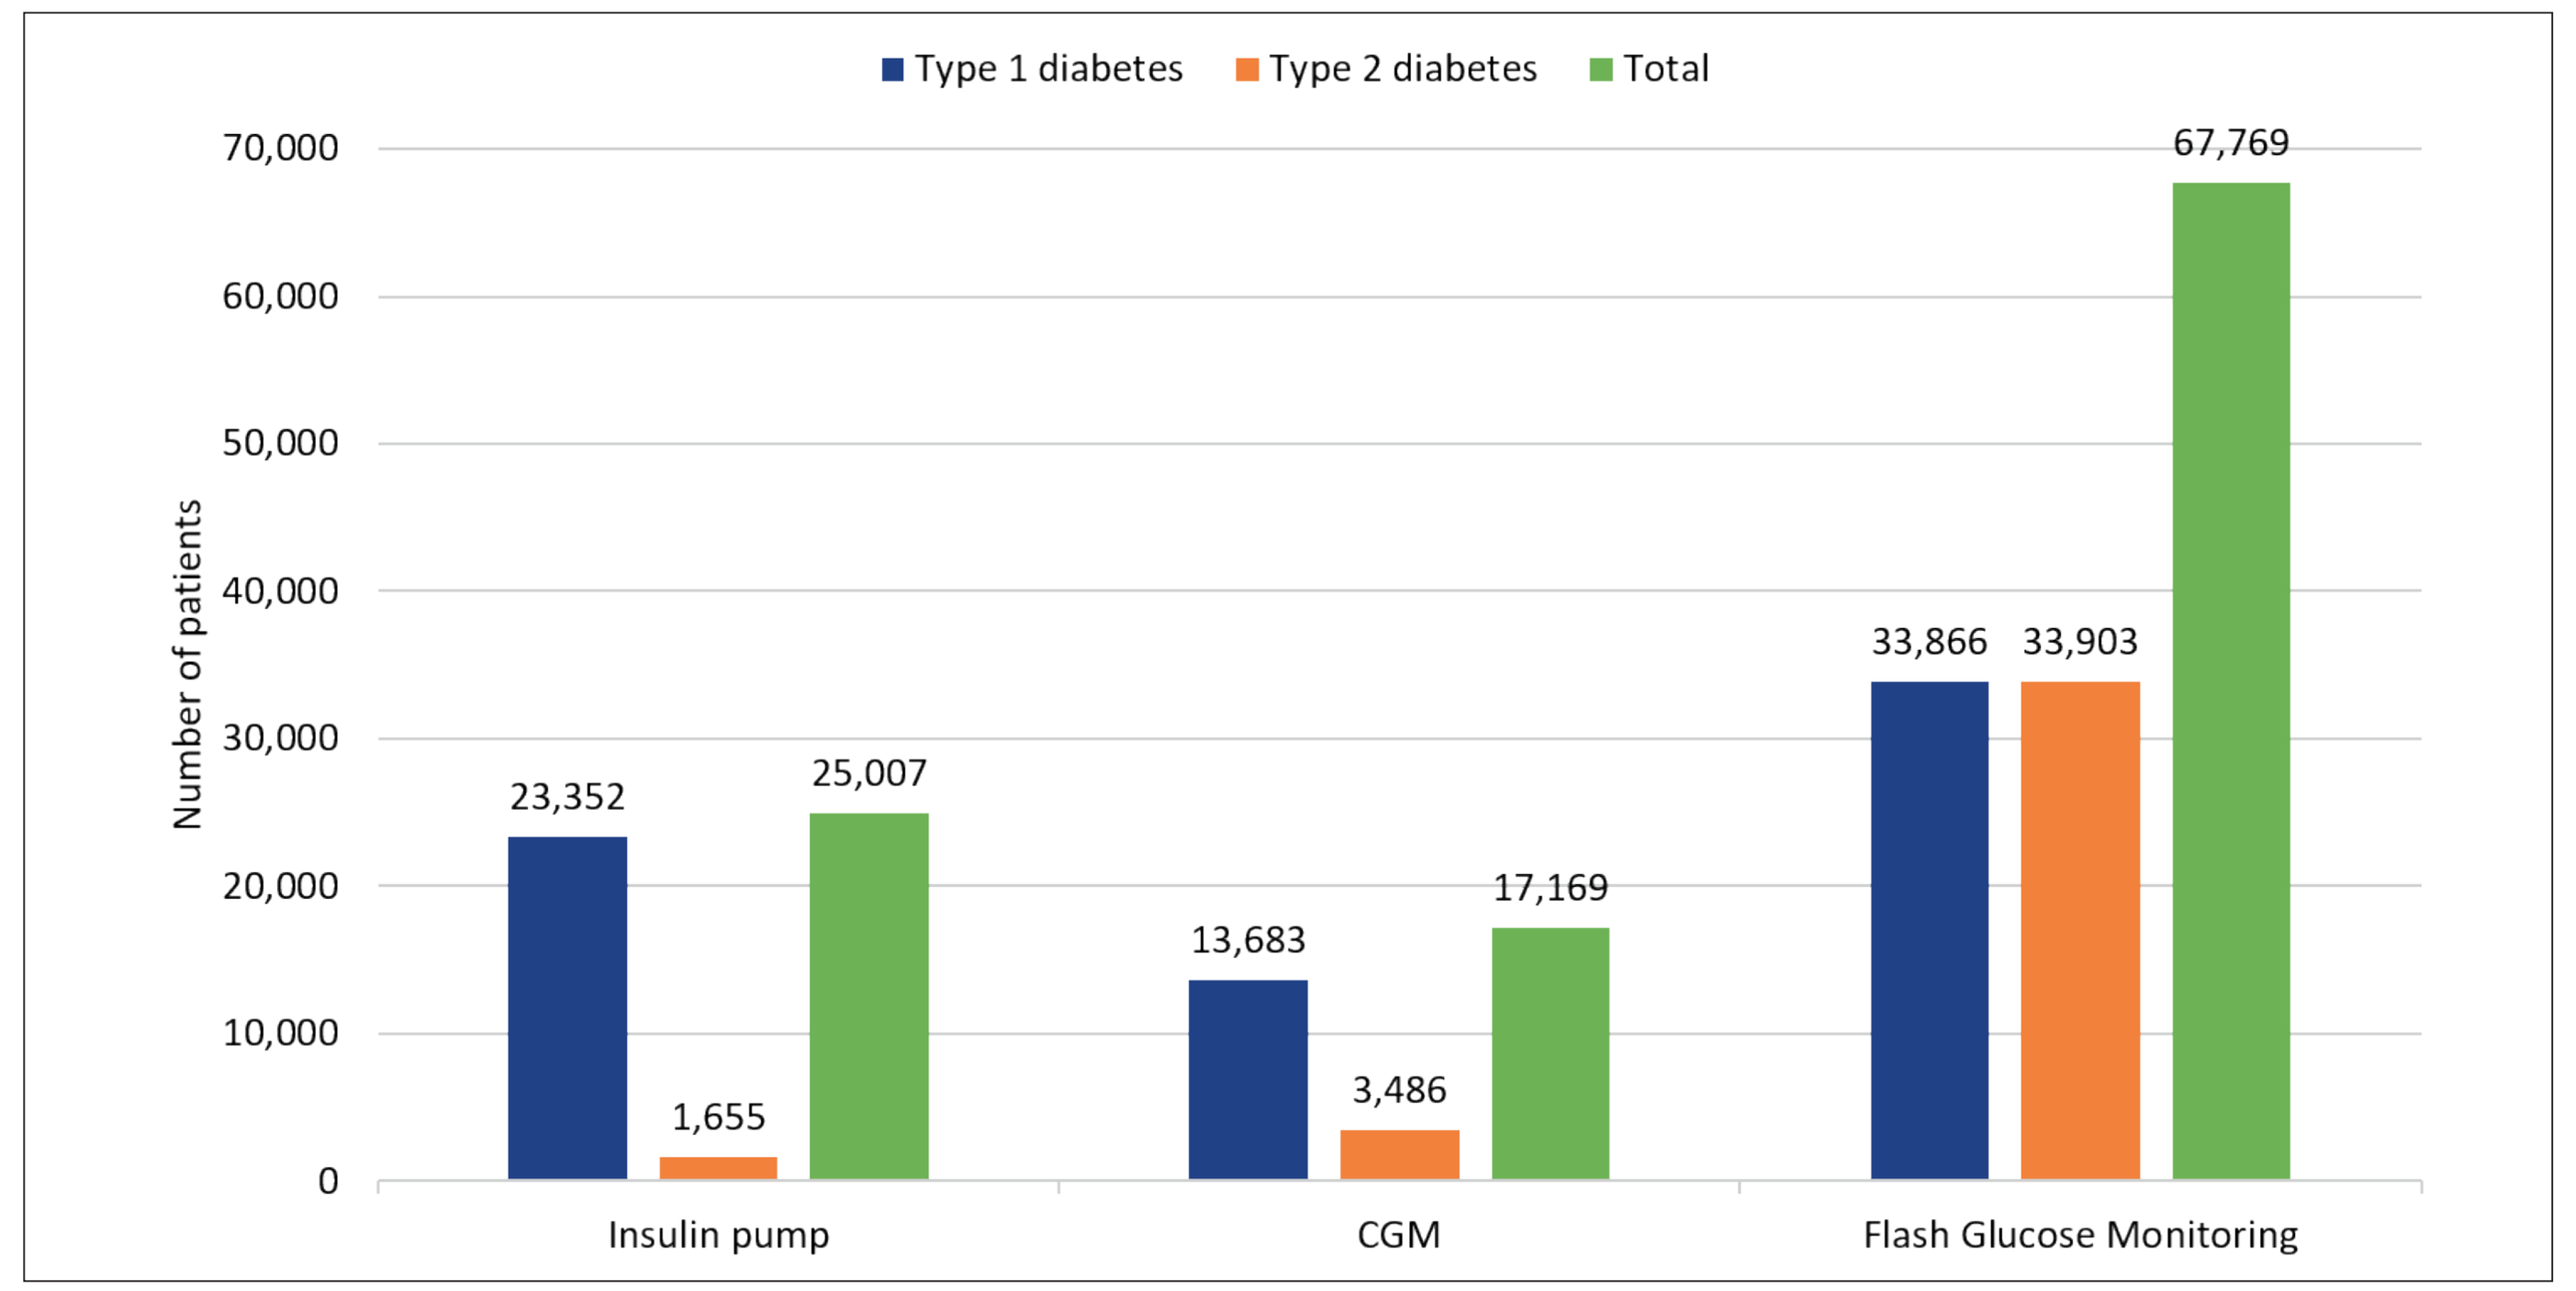

Type 1 diabetes
Type 2 diabetes
Total
Number of patients
70,000
60,000
50,000
40,000
30,000
20,000
10,000
0
23,352
1,655
25,007
13,683
3,486
17,169
33,866
33,903
67,769
Insulin pump
CGM
Flash Glucose Monitoring

Figure 1: Physician's estimate of the number of patients with insulin pumps or continuous glucose monitoring (CGM) systems

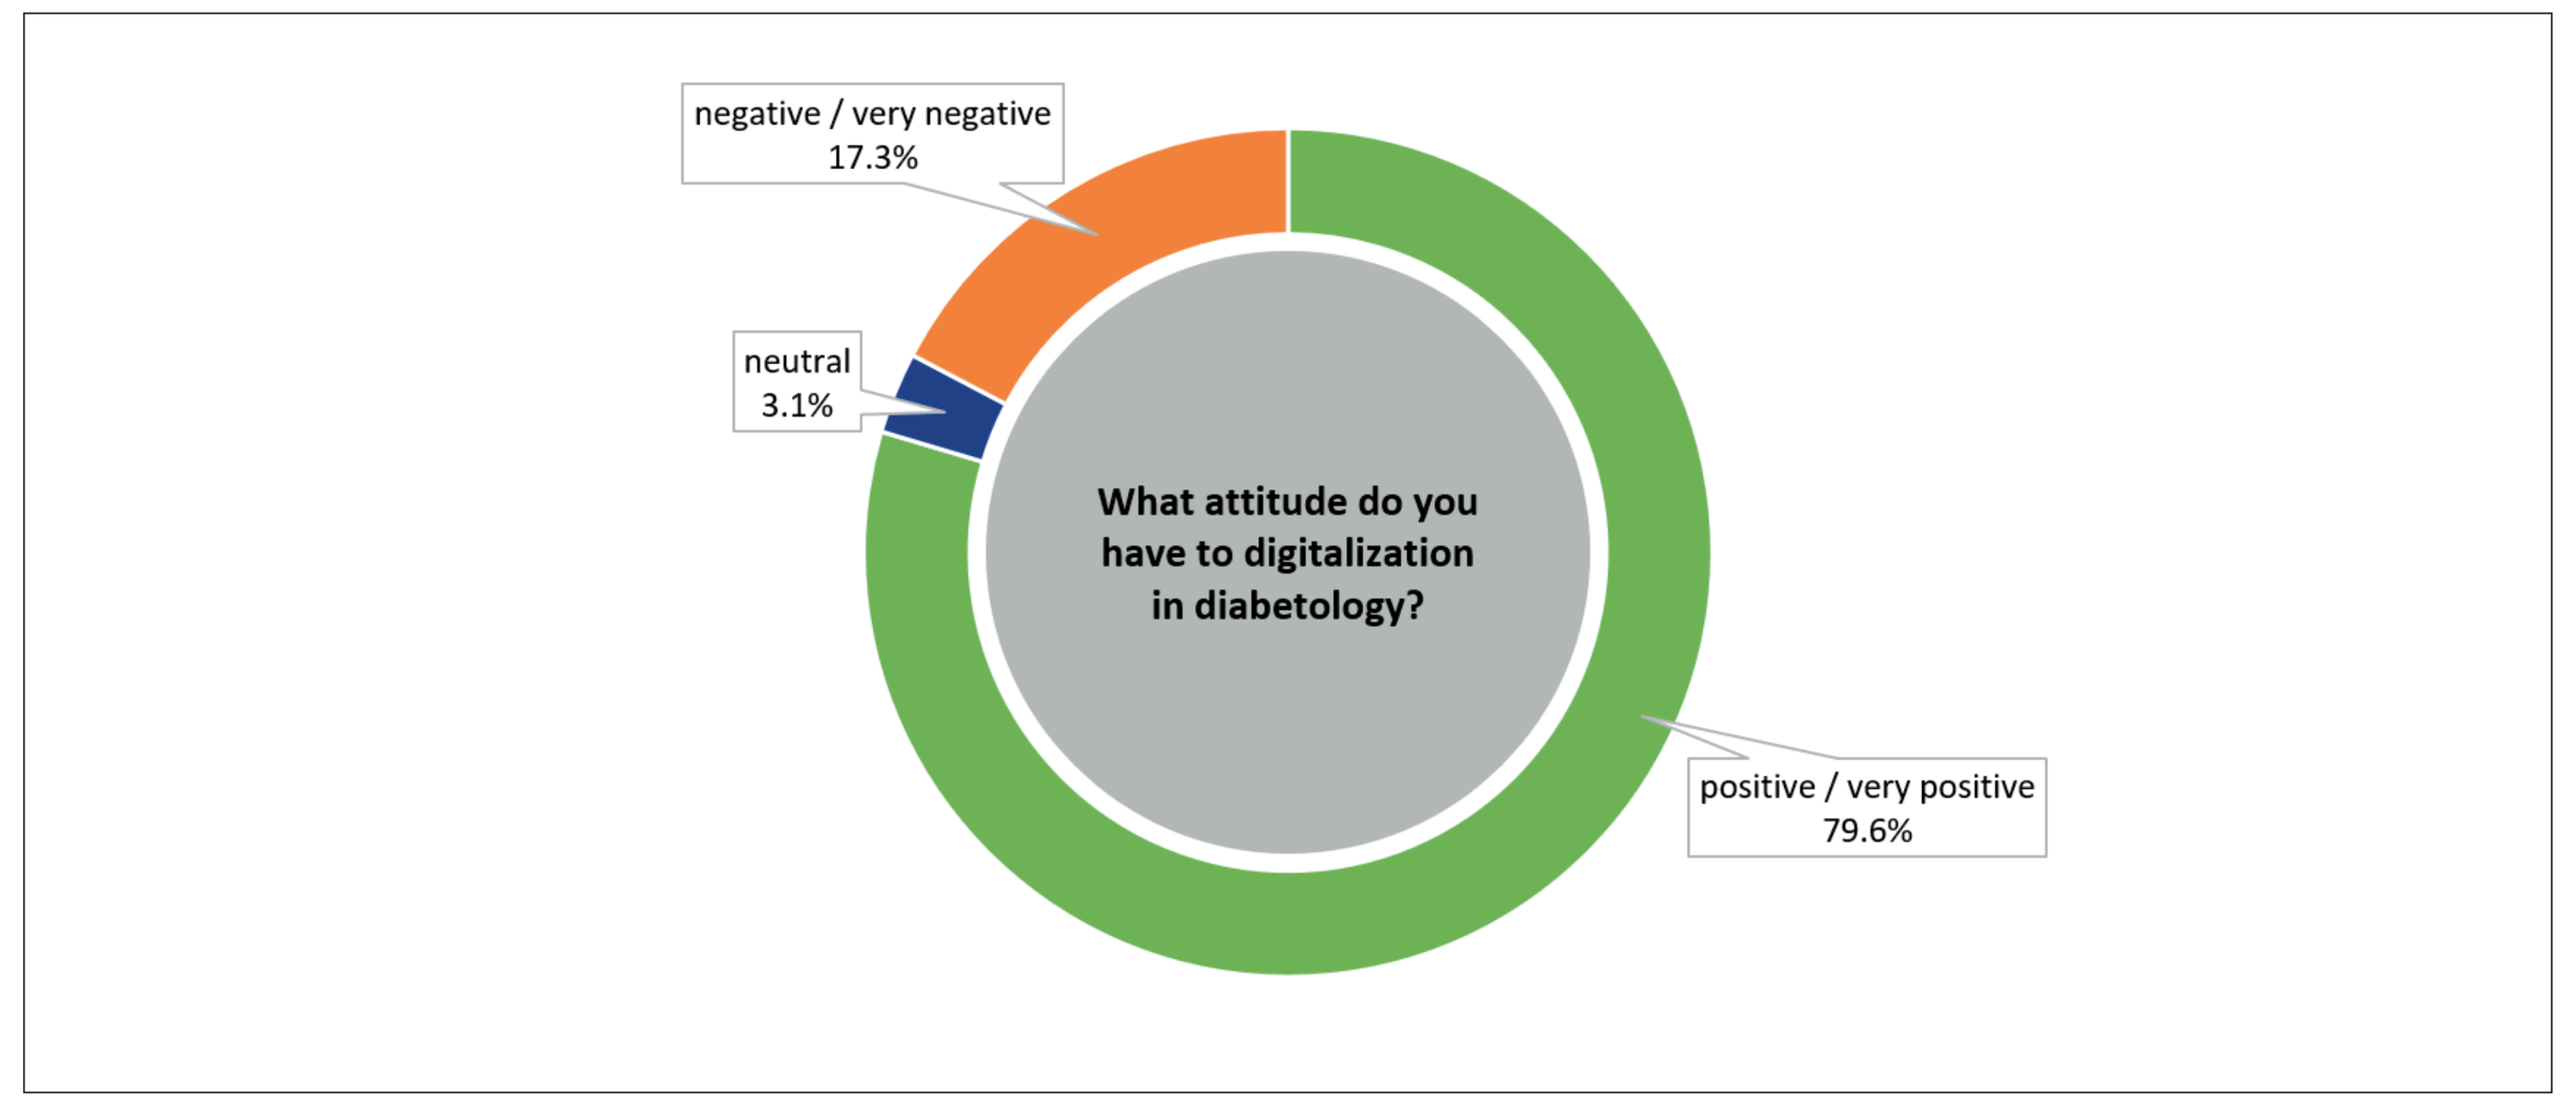

negative / very negative
17.3%
neutral
3.1%
What attitude do you have to digitalization in diabetology?
positive / very positive
79.6%

Figure 2: Attitude towards digitalization in diabetology (in %)

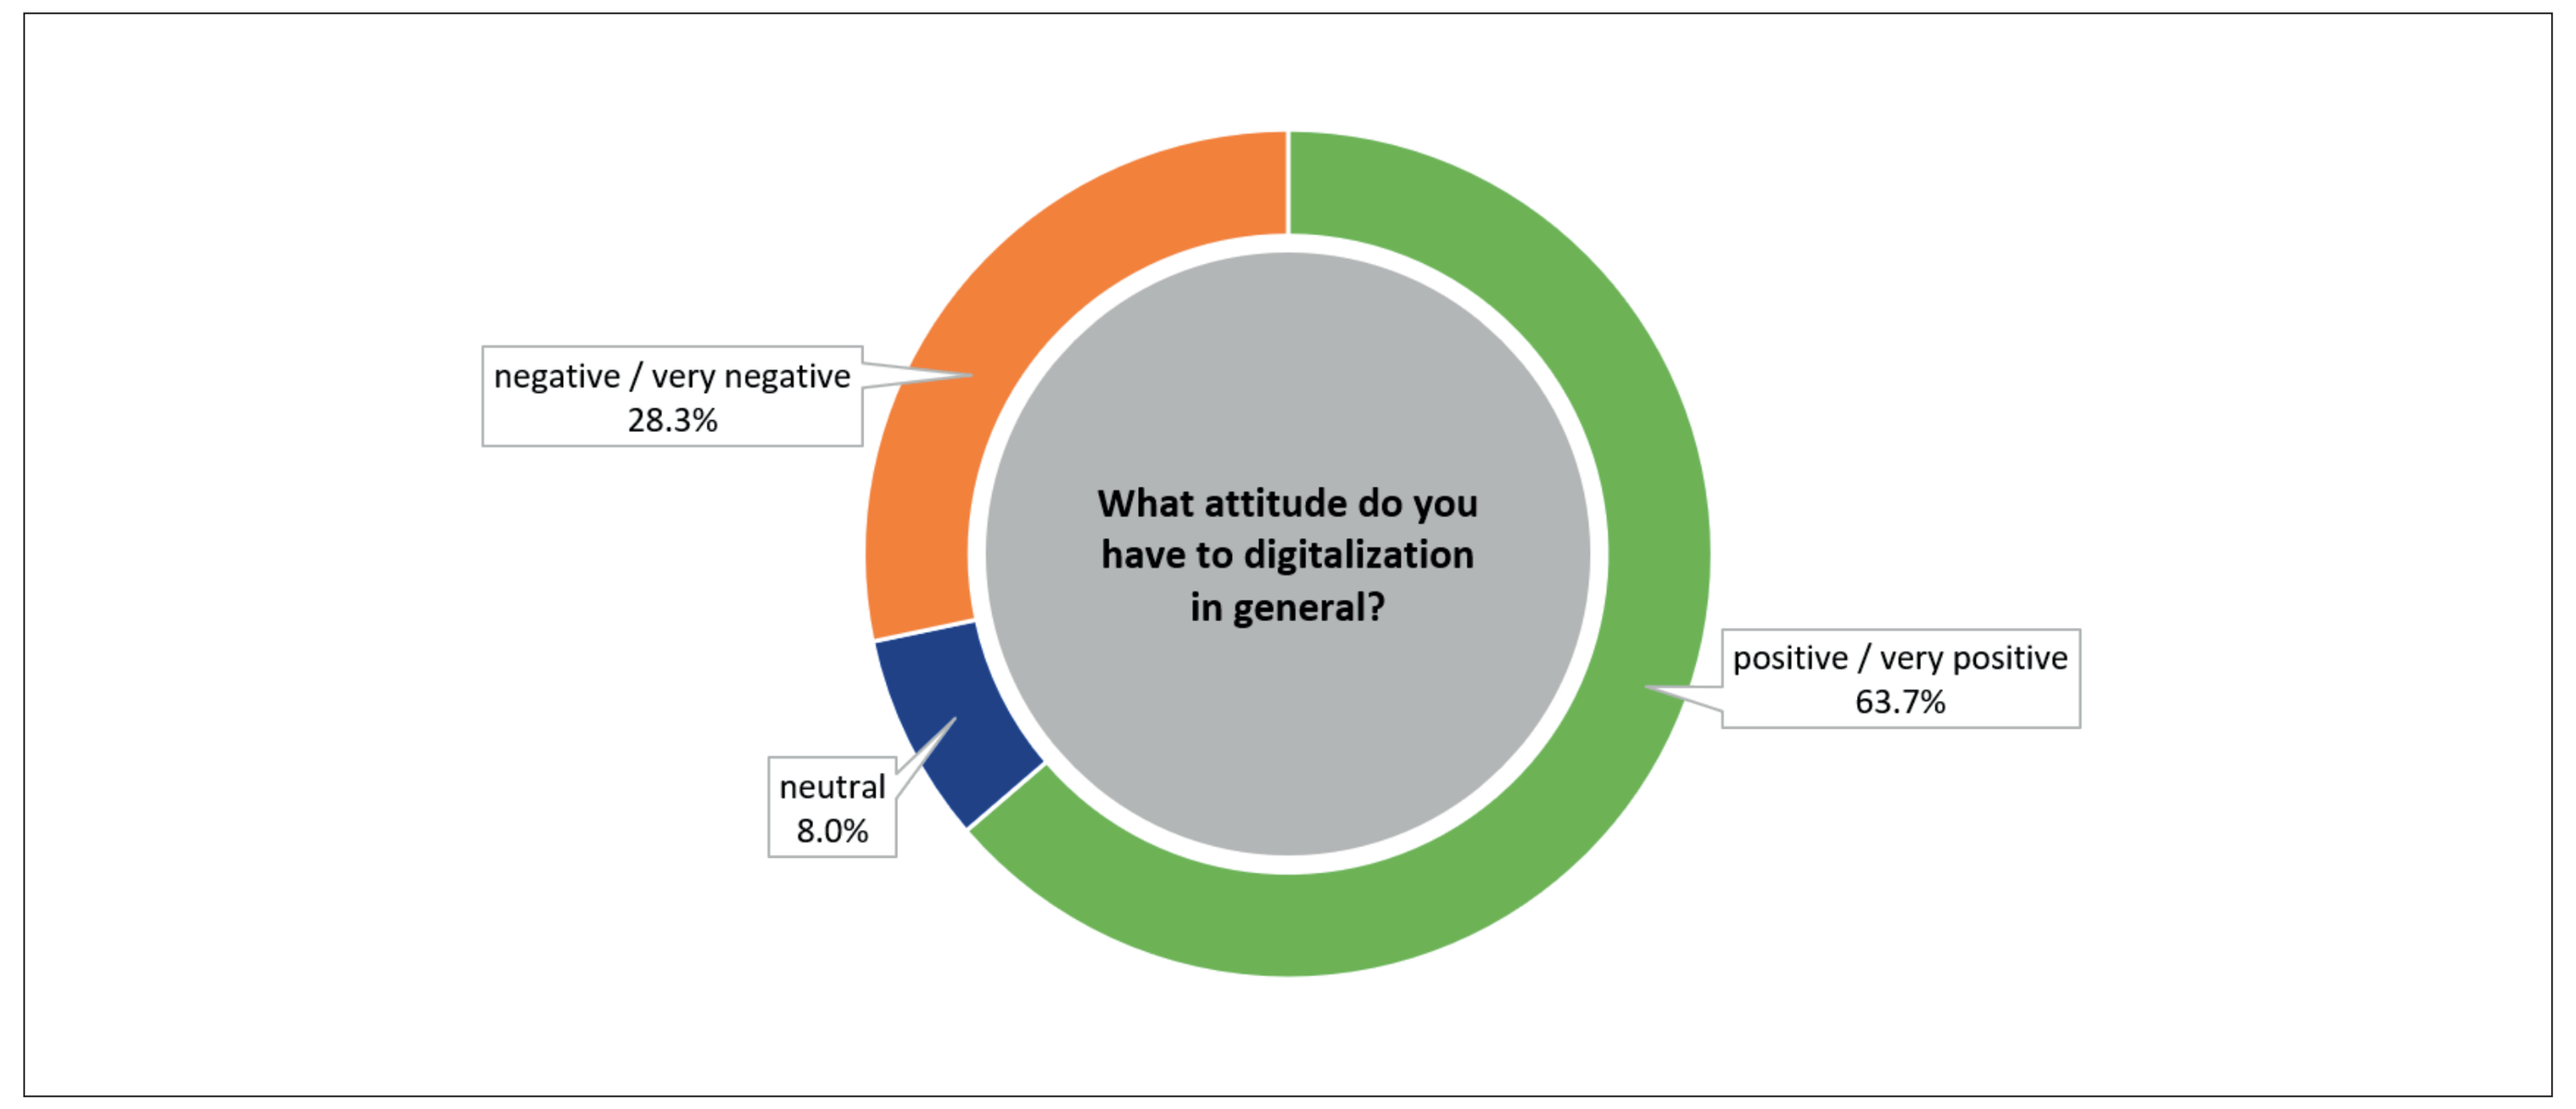

negative / very negative
28.3%
neutral
8.0%
What attitude do you have to digitalization in general?
positive / very positive
63.7%

Figure 3: Attitude towards digitalization in general (in %)

Contact Information

FIDAM - Research Institute Diabetes Academy Mergentheim
Bernhard Kulzer
97980 Bad Mergentheim
Germany
Phone: +49 7931 594-151
kulzer@fidam.de

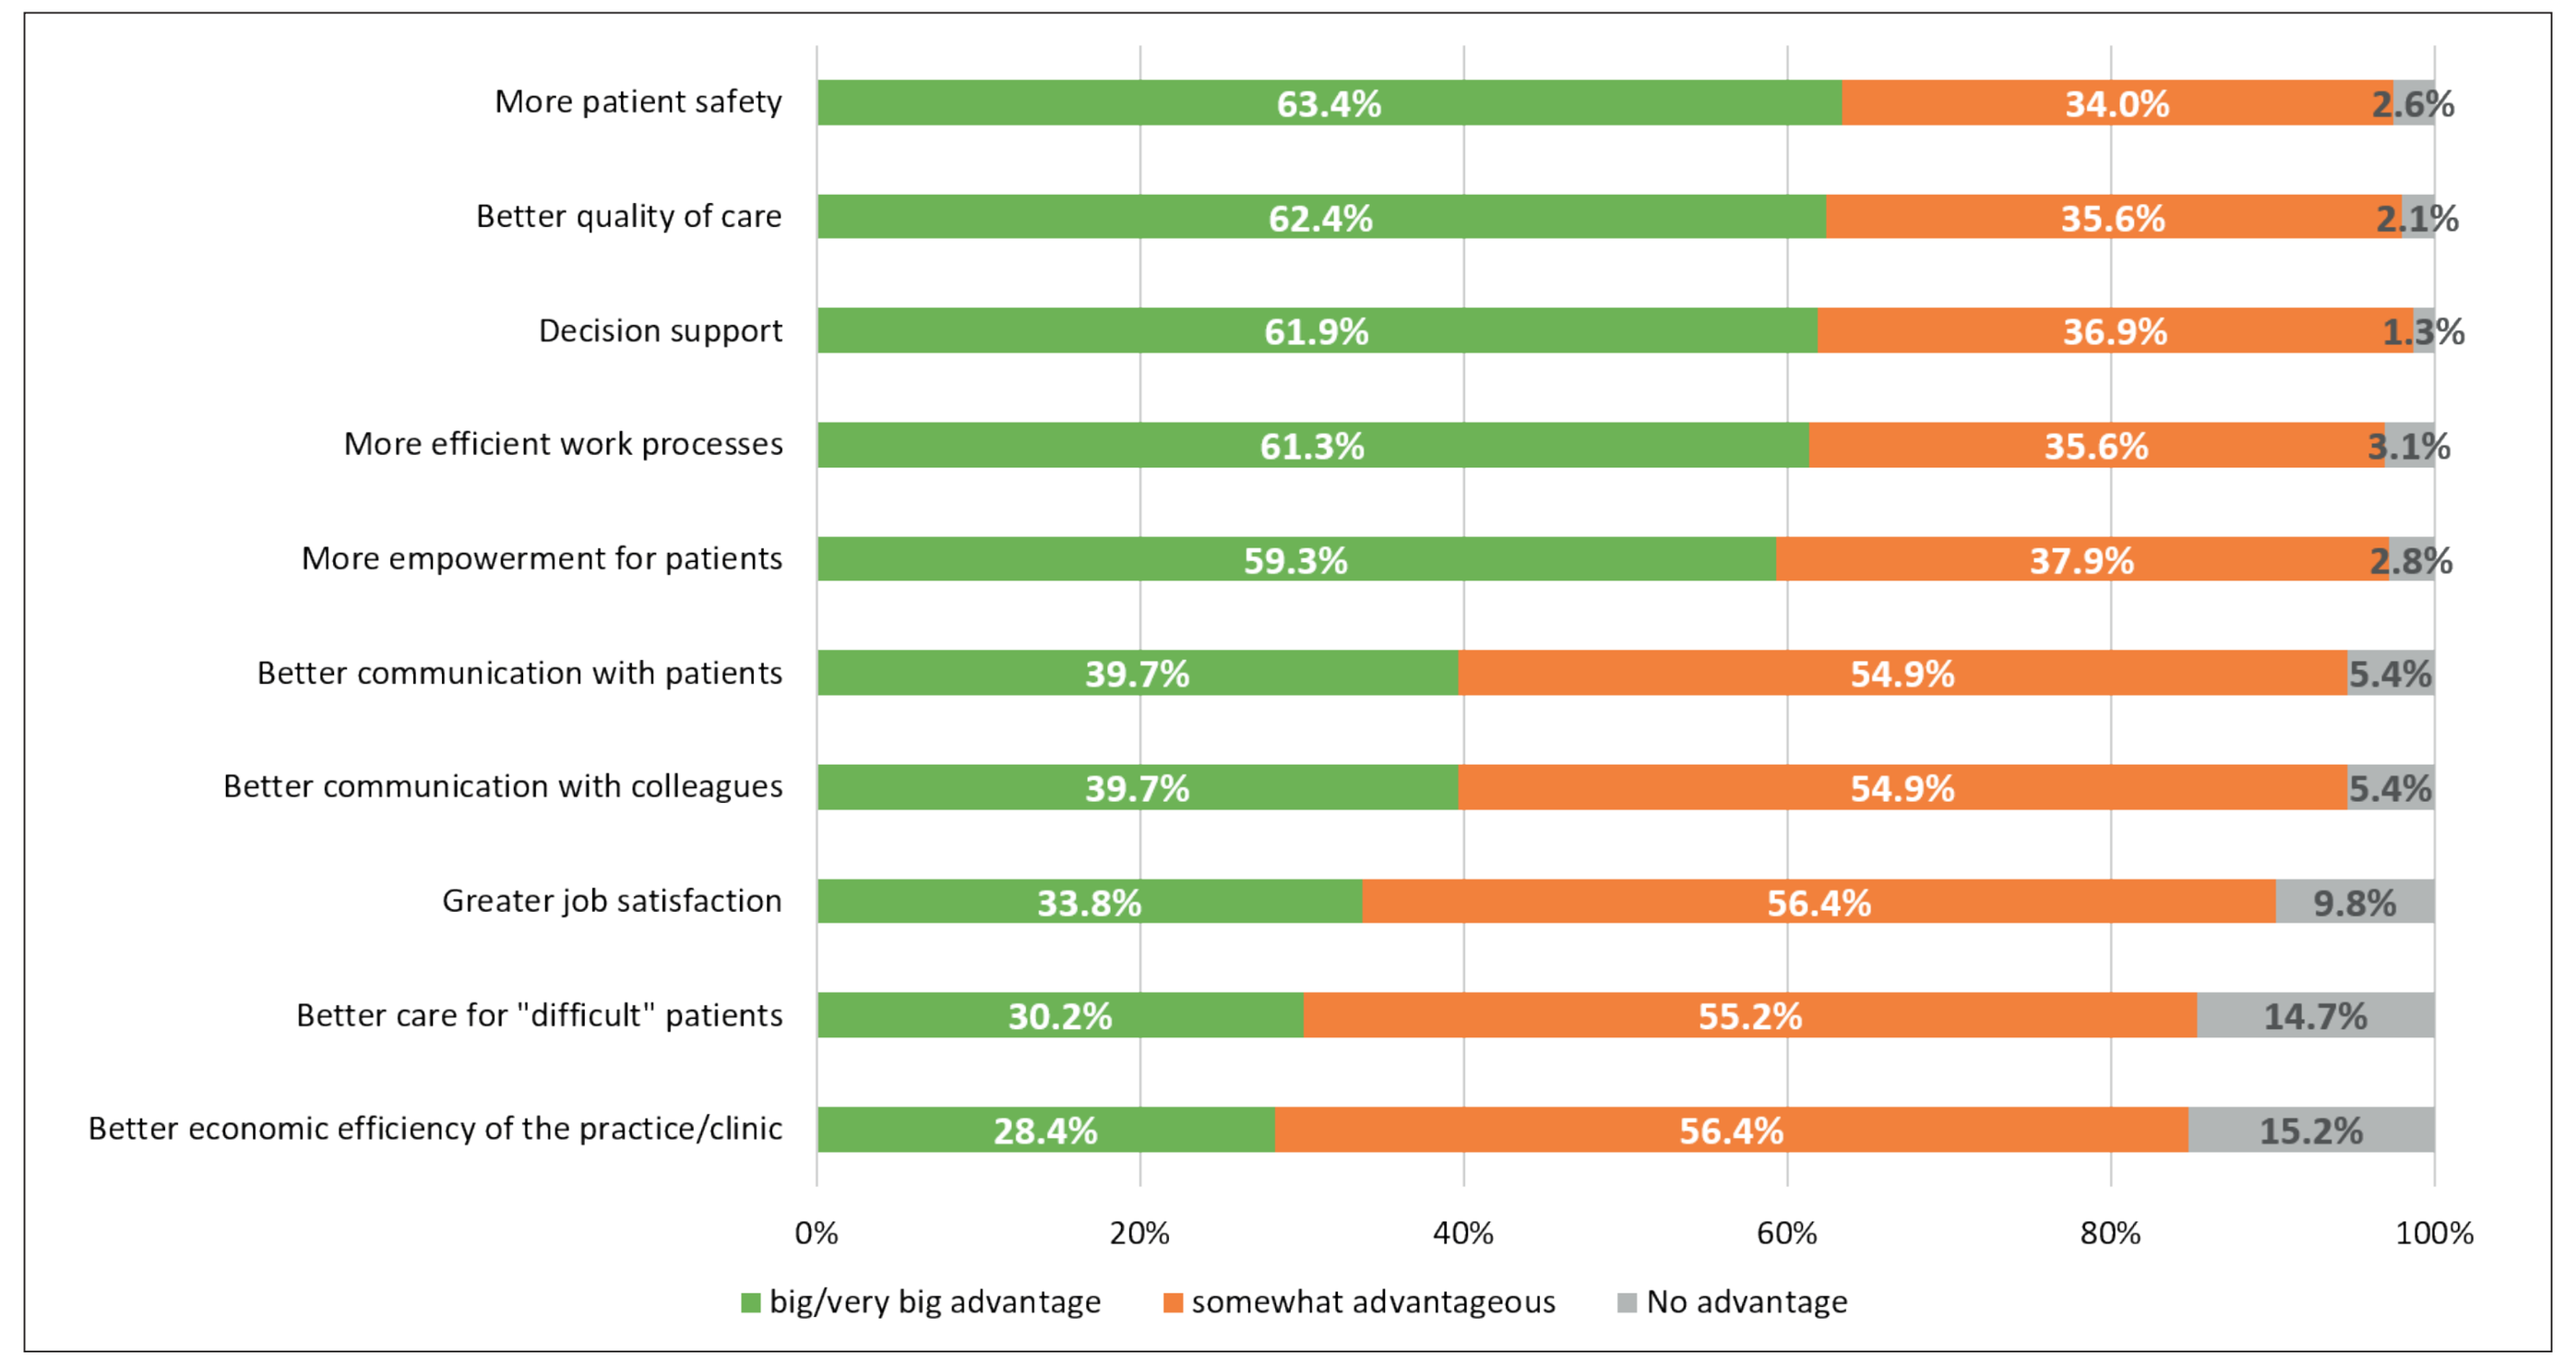

More patient safety 63.4% 34.0% 2.6%
Better quality of care 62.4% 35.6% 2.1%
Decision support 61.9% 36.9% 1.3%
More efficient work processes 61.3% 35.6% 3.1%
More empowerment for patients 59.3% 37.9% 2.8%
Better communication with patients 39.7% 54.9% 5.4%
Better communication with colleagues 39.7% 54.9% 5.4%
Greater job satisfaction 33.8% 56.4% 9.8%
Better care for "difficult" patients 30.2% 55.2% 14.7%
Better economic efficiency of the practice/clinic 28.4% 56.4% 15.2%
0% 20% 40% 60% 80% 100%
big/very big advantage
somewhat advantageous
No advantage

Figure 4: Physician's appraisal of advantages of digitalization

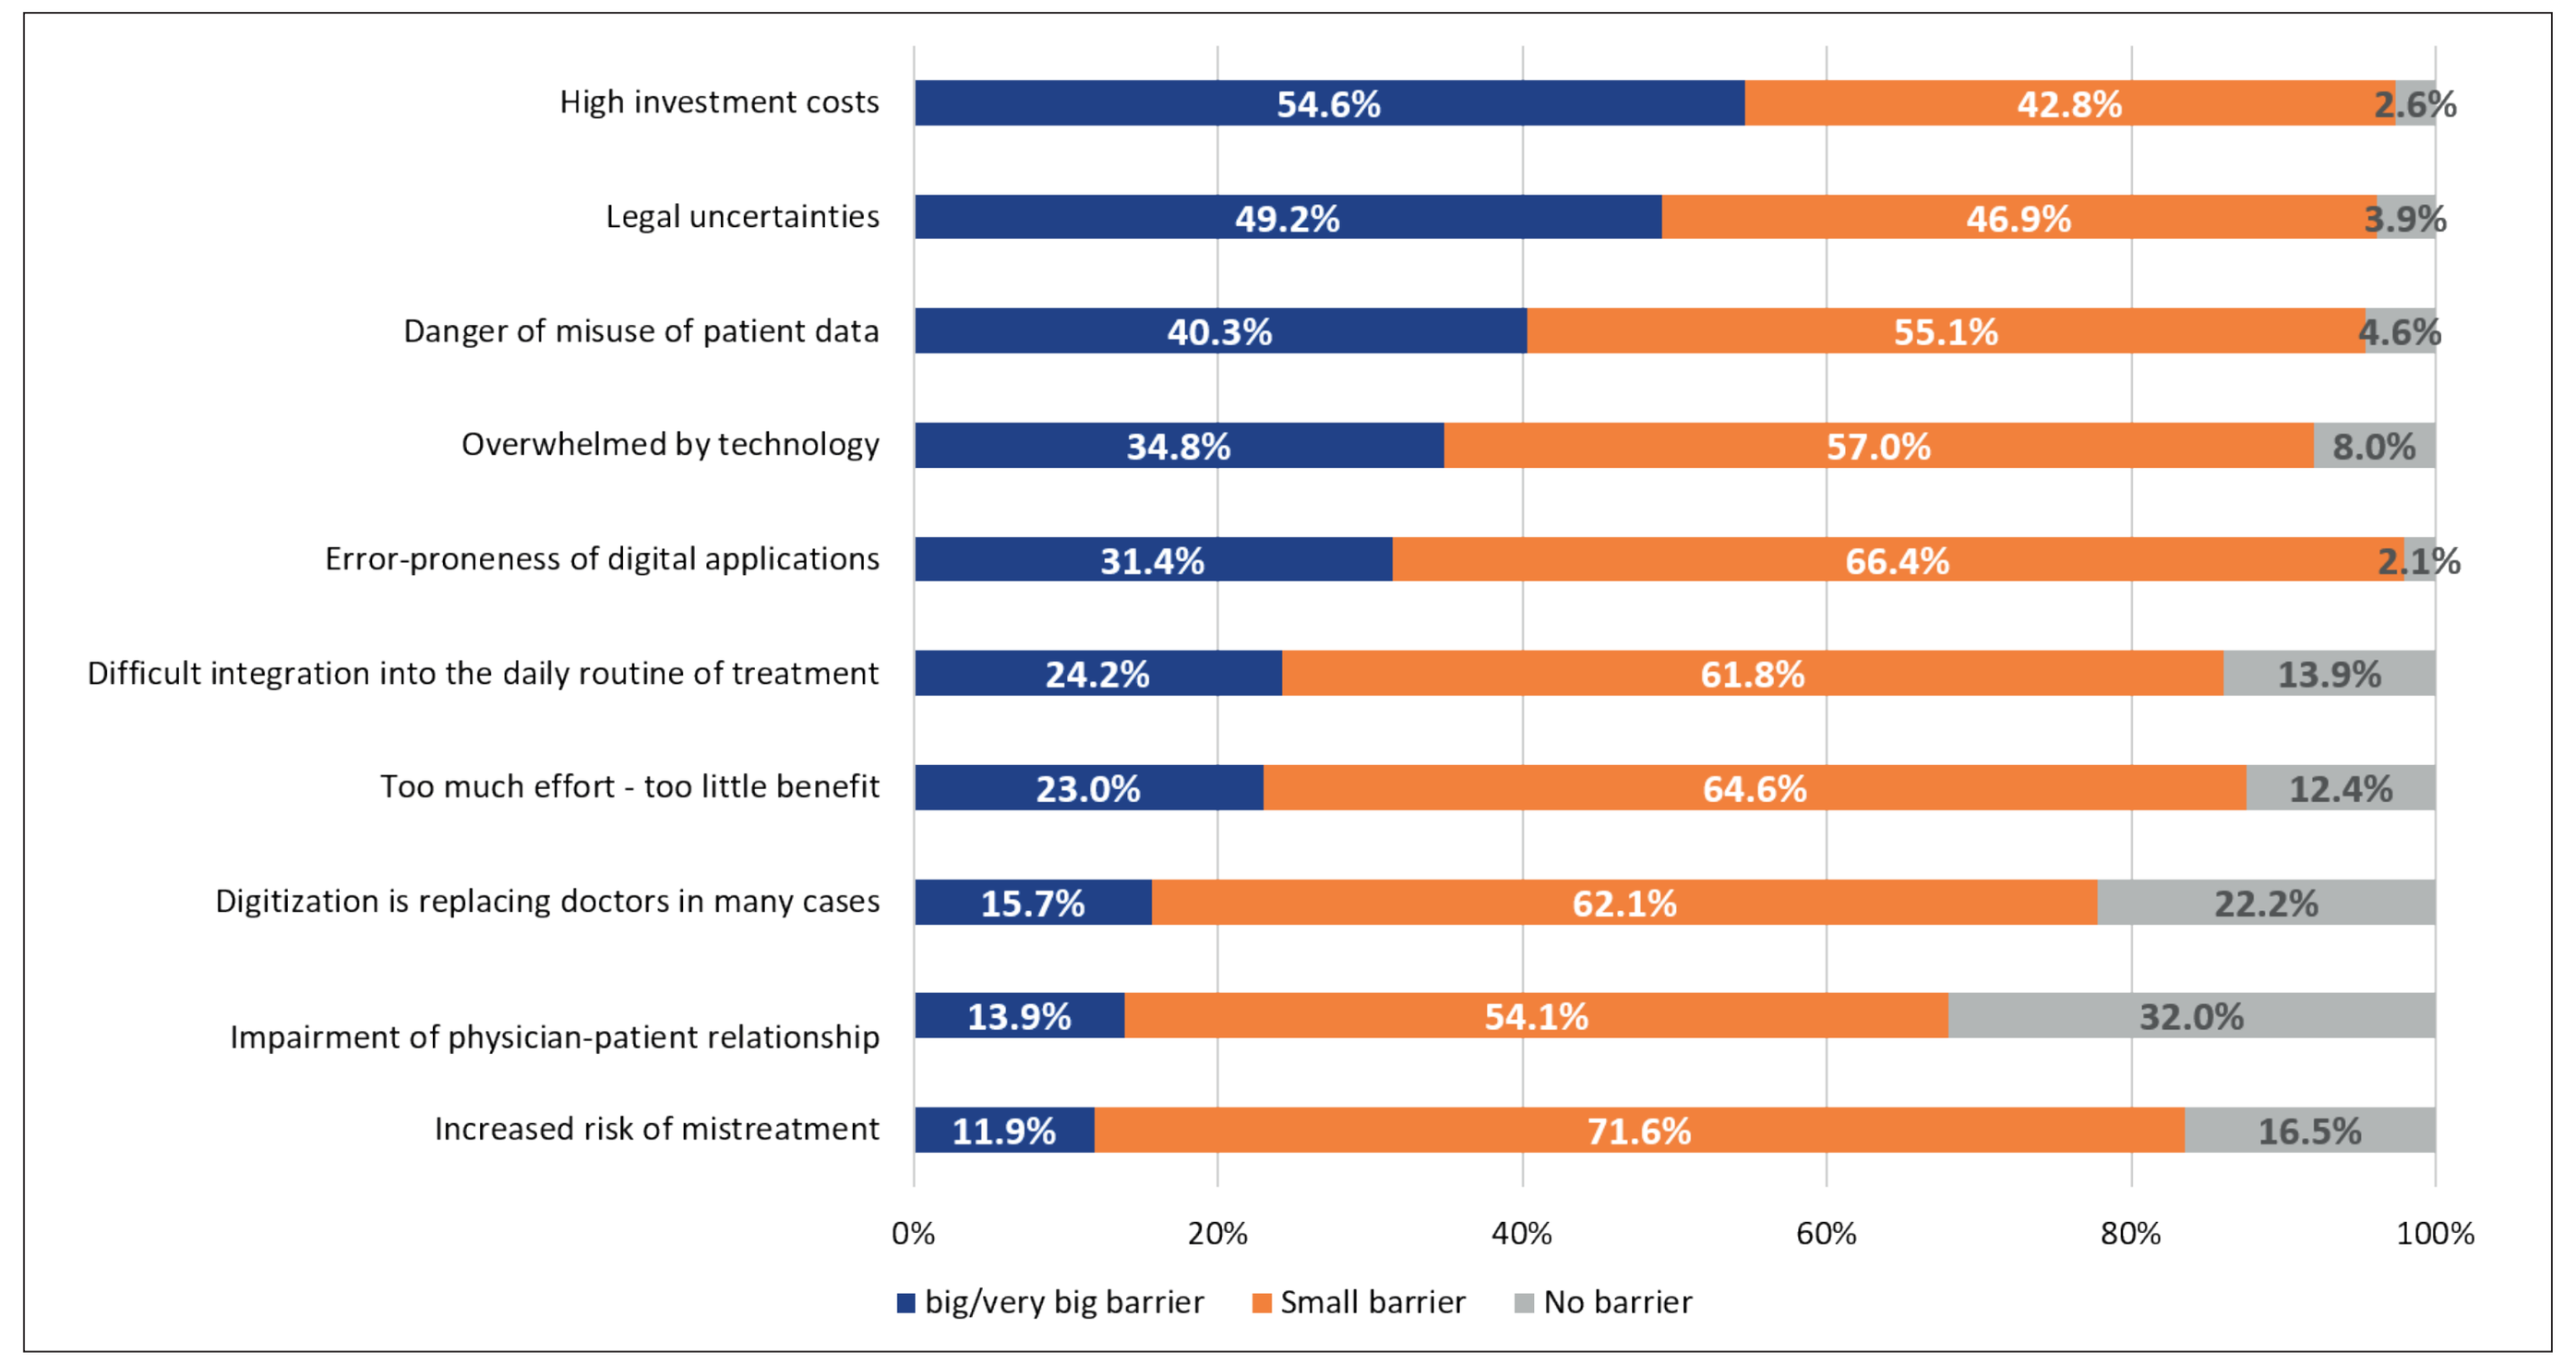

High investment costs 54.6% 42.8% 2.6%
Legal uncertainties 49.2% 46.9% 3.9%
Danger of misuse of patient data 40.3% 55.1% 4.6%
Overwhelmed by technology 34.8% 57.0% 8.0%
Error-proneness of digital applications 31.4% 66.4% 2.1%
Difficult integration into the daily routine of treatment 24.2% 61.8% 13.9%
Too much effort - too little benefit 23.0% 64.6% 12.4%
Digitization is replacing doctors in many cases 15.7% 62.1% 22.2%
Impairment of physician-patient relationship 13.9% 54.1% 32.0%
Increased risk of mistreatment 11.9% 71.6% 16.5%
0% 20% 40% 60% 80% 100%
big/very big barrier
Small barrier
No barrier

Figure 5: Physician's appraisal of barriers towards digitalization

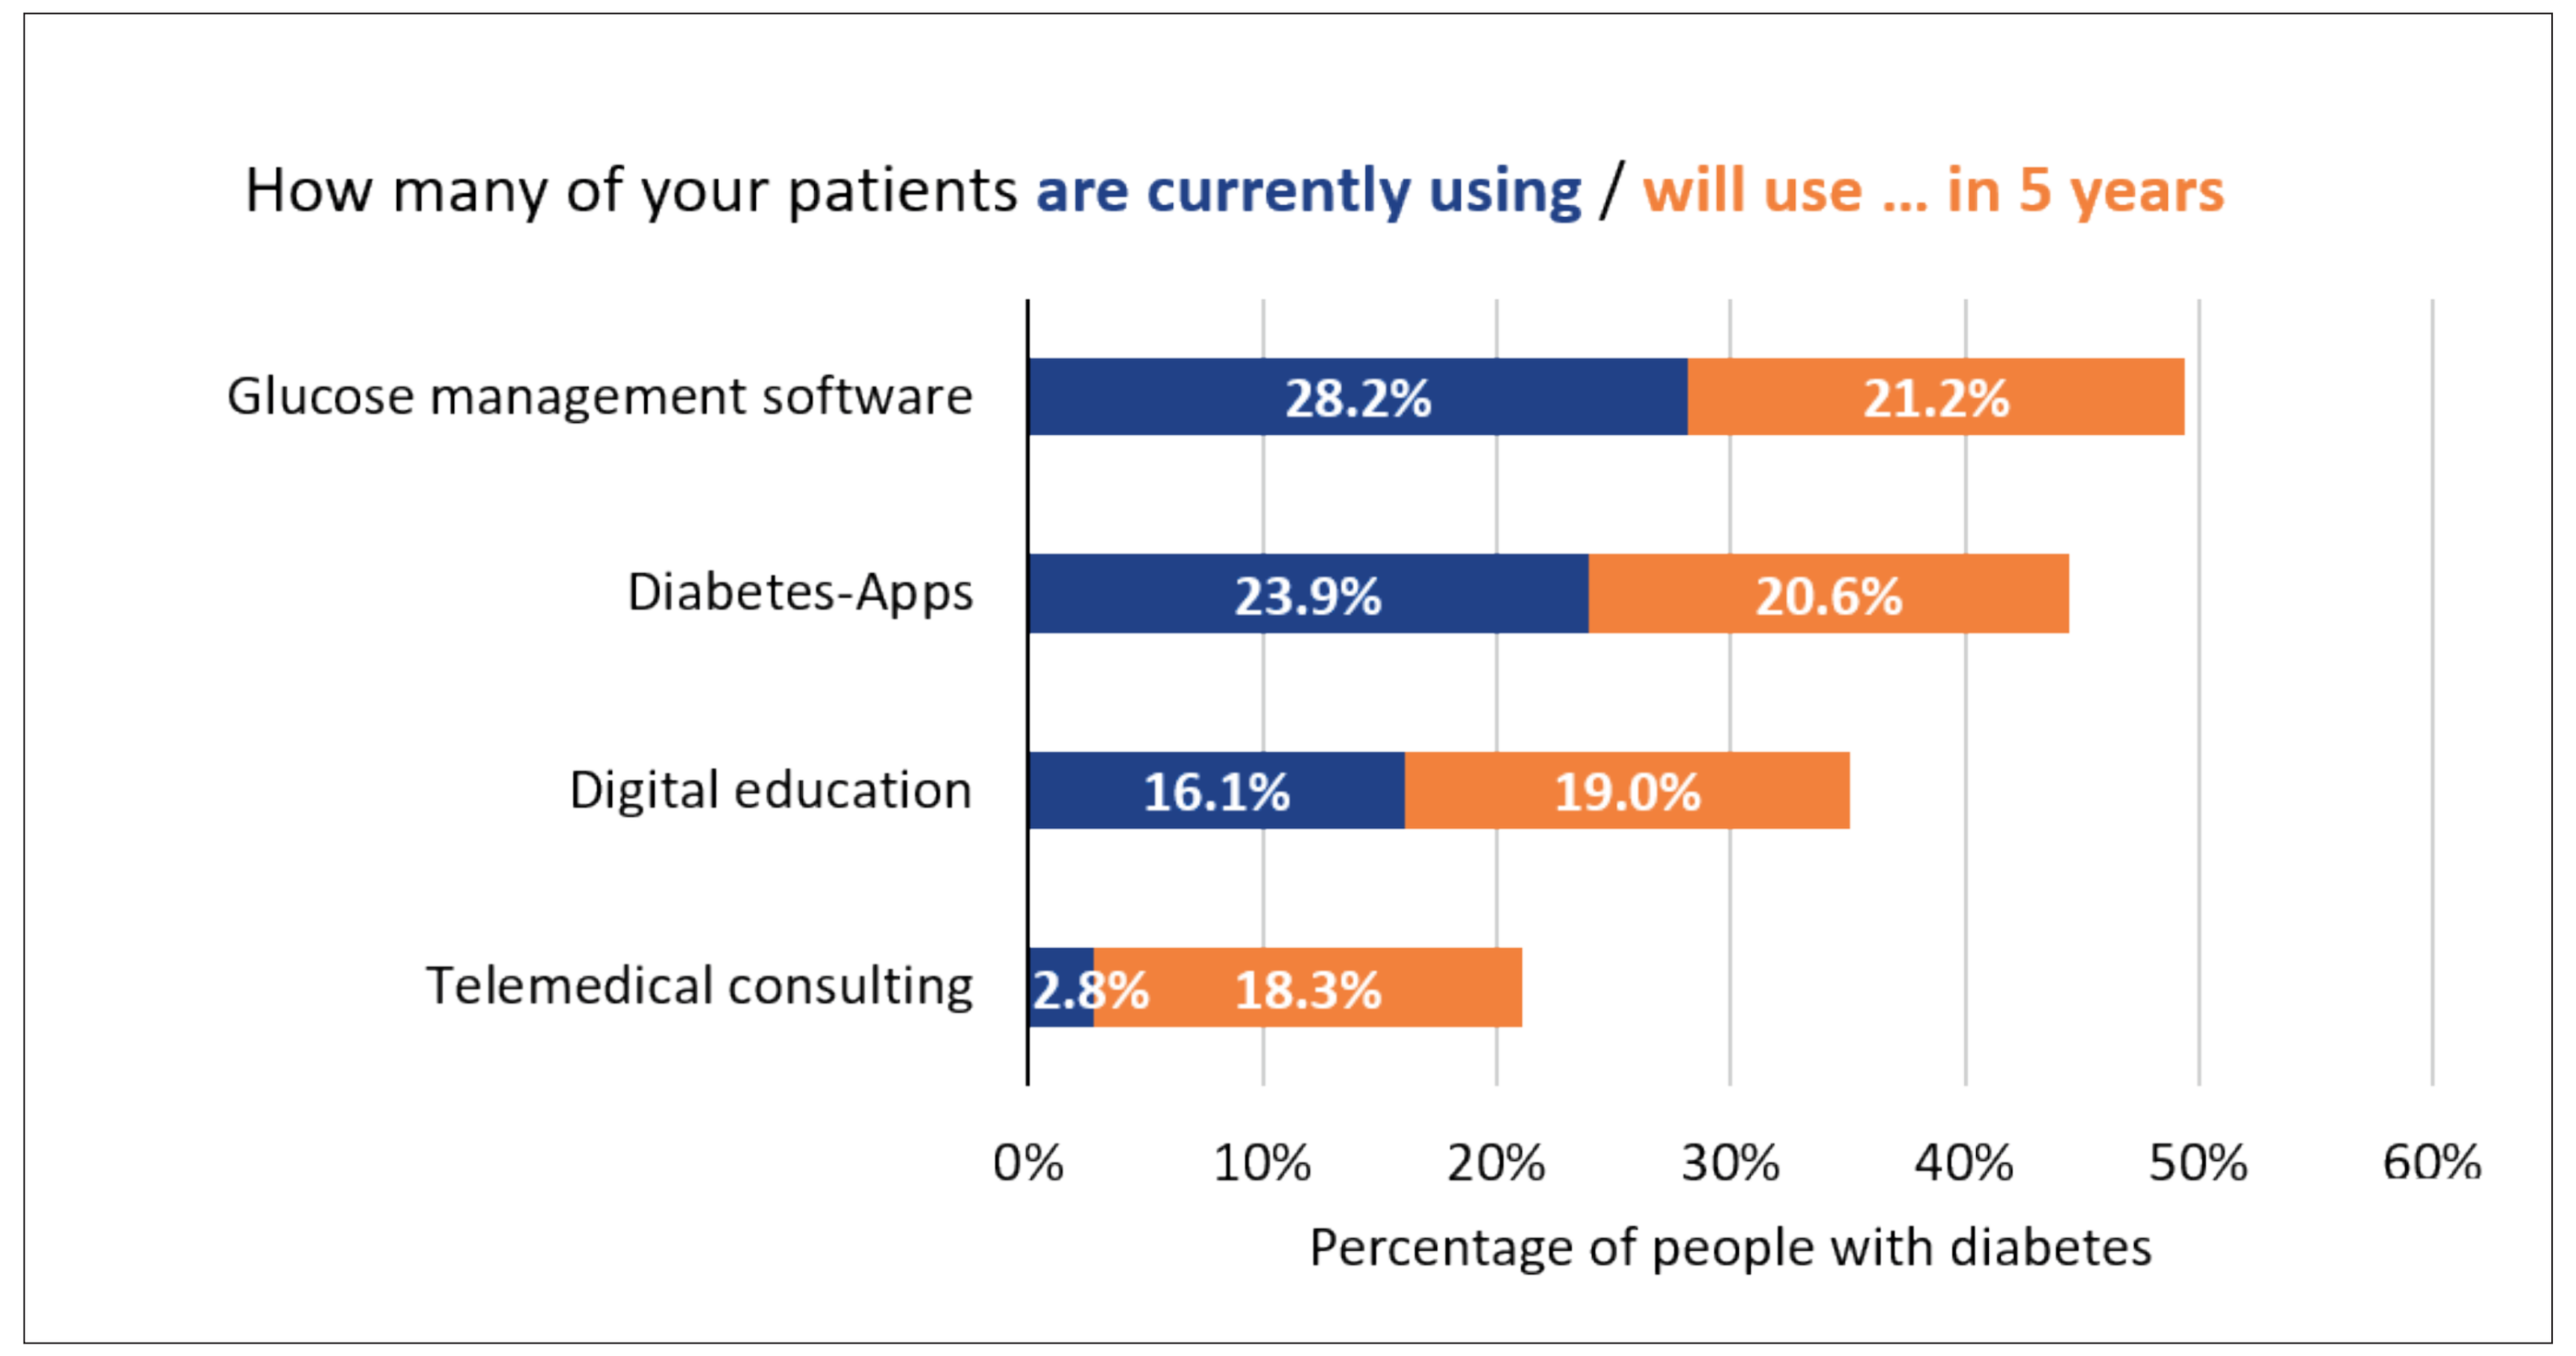

How many of your patients are currently using / will use ... in 5 years
Glucose management software 28.2% 21.2%
Diabetes-Apps 23.9% 20.6%
Digital education 16.1% 19.0%
Telemedical consulting 2.8% 18.3%
0% 10% 20% 30% 40% 50% 60%
Percentage of people with diabetes

Figure 6: Physician's appraisal of current and future use of digital possibilities